Population Pharmacokinetics (PPK) and Pharmacokinetic-Pharmacodynamic (PK/PD) of Vicriviroc in Treatment Naïve HIV Patients

Christine Xu,[1] Honghong Zhou,[2] Gopal Krishna,[1] Michael McCarthy[3]

[1]Clinical PK/PD, Department of Drug Metabolism and Pharmacokinetics, [2]BARDS,
[3]Global Clinical Research, Merck Research Labs, Kenilworth, NJ USA

MERCK

Abstract

Objectives: Vicriviroc is a potent CCR5 antagonist for treating HIV-1 infection. This study aimed to (1) assess vicriviroc pharmacokinetic (PK) profile and interpatient versus intrapatient variability in HIV positive patients using PPK, and (2) to explore PK/PD association using drug exposure and efficacy.

Methods: This global Phase 2 trial explored a class-sparing regimen of vicriviroc, a CCR5 antagonist vs Truvada, each in combination with ritonavir-boosted atazanavir (ATV/r). CCR5 HIV-infected treatment-naïve subjects were randomized 1:1 to open-label treatments with 95 and 123 subjects in Stage 1 and 2, respectively. The PPK analysis was performed using NONMEM based on plasma samples from patients receiving vicriviroc. Two plasma samples were collected pre-dose and approximately 1 hr post-dose on Weeks 4 and 12. Since the sparse PK sampling scheme was utilized in this Phase 2 trial, PK data from Phase I studies were combined and used for the population PK analysis. The influence of demographic and clinical characteristics on clearance and volume of distribution were examined. The drug exposure (AUC, Cmin and Cmax) was estimated for each patient and the exposure-virologic response association was explored.

Results: 105 vicriviroc treated patients contributed to 402 vicriviroc concentrations. Two-compartment model with first-order absorption and elimination was chosen as the pharmacokinetic base model. The apparent clearance (CL/F) was 3.39 L/h, apparent volume of distribution of the central compartment (Vc/F) was 170 L, and the absorption rate constant (Ka) was 0.743 hr-1. Of the covariates evaluated, body weight was a significant covariate for CL/F and age was a significant covariate for Vc/F. A large interpatient variability was found for Ka (CV 70%), while the intrapatient variability was relatively small (CV 19%). The final model was evaluated by the bootstrap technique and the visual predictive check. The correlation between trough drug concentration (Cmin) and viral load change was assessed. Higher response rates were observed in naive subjects with higher vicriviroc Cmin.

Conclusions: A two-compartment model adequately described the vicriviroc PK in naïve HIV patients. Body weight and age were significant covariates. The integrated population PK model and PK-PD association can be used to predict antiviral activity and select the optimal dose regimen in naive patients.

Background

- Human immunodeficiency virus (HIV) has evolved from a short, fatal disease to a chronic, manageable disease requiring lifelong therapy.
- Vicriviroc (SCH 417690) is a potent CCR5 receptor antagonist, which belongs to the new class of HIV entry inhibitors.
- This class is designed to block cell surface receptors, such as CCR5 or CXCR4, as well as compounds that block HIV fusion with the cell surface. Unlike existing HIV drugs that work inside the cell and target viral enzymes involved in the replication of the virus, entry inhibitors block HIV before the virus enters the cell.
- Vicriviroc provides anti-HIV activity similar to that of dual-nucleoside treatment (~1.6 $\log_{10}$ decline in HIV RNA copies/mL).
- A regimen of VCV plus ritonavir-boosted atazanavir (ATV/r) could provide a well-tolerated, convenient HIV treatment regimen with efficacy equivalent to that of nucleoside therapy, while sparing patients from the toxicities associated with this class of drugs.

Study Design

- A Phase II, randomized, open-label, comparative study conducted in the United States, European Union, Latin America, and South Africa
- Target population size was approximately 200 patients enrolled in 2 stages
 - Stage 1: Approximately 80 patients
 - Stage 2: Approximately 120 patients
- Enrollment in Stage 2 was performed after 24-week data from Stage 1 were reviewed by an external, independent data and safety monitoring board
- Patients were randomly selected in a 1:1 ratio to receive
 - VCV 30 mg QD + ATV/r 300 mg/100 mg QD
 - Tenofovir/Emtricitabine 300 mg/200 mg QD + ATV/r 300 mg/100 mg QD
- Primary endpoint: Change from baseline HIV RNA
 - Key secondary endpoint: % <50 copies/mL at 48 weeks (later amended to continue to 96 weeks)
 - Other secondary endpoints: virologic failure, viral loads in patients with virologic failure, and change in CD4 count from baseline

Objectives

(1) To assess vicriviroc pharmacokinetic (PK) profile and interpatient versus intrapatient variability in HIV positive patients using PPK, and

(2) To explore PK/PD association using drug exposure and efficacy.

Methods

Data

Data Inclusion and Exclusion Criteria

- The following data inclusion criteria were employed in assembling of data from Phase I and Phase II studies:
 - Subjects dosed with vicriviroc in combination with a ritonavir-containing regimen
 - Vicriviroc concentrations at steady-state
 - Subjects dosed with tablet or capsule formulations of vicriviroc
 - Subjects given dose of vicriviroc at 30 mg
- The following data exclusion criteria were used:
 - Vicriviroc concentrations below limit of quantification
 - PK samples without dosing record prior to PK sampling
 - PK samples without sampling record

PK Database

- Since the sparse PK sampling scheme was utilized in Phase II study, data from 3 Phase I studies were combined and used for the population PK analysis to estimate the pharmacokinetic parameters for HIV infected subjects enrolled in Phase II study.

Population PK Analysis

- The population PK analysis was conducted using the pooled data from three Phase 1 studies and one Phase 2 study.
- Population PK analysis was conducted via nonlinear mixed-effect model approach using NONMEM program. The first-order conditional estimation method with interaction (FOCE-I) was employed during the model development, assuming that the convergence of minimization process is successful.
- Covariates included in the population PK dataset were age, body weight, gender, ethnicity, alkaline phosphatase (ALP), AST (SGOT), ALT (SGPT), total bilirubin, serum creatinine concentration and disease status. Creatinine clearance (CrCL) was calculated using Cockcroft-Gault equation. If the calculated creatinine clearance was >150 mL/min, the value of creatinine clearance was entered as 150 mL/min.

Base Model

- Exploratory data analysis included checking of PK and covariate outliers, and examining the distributions and correlations of the covariates.
- With the prior knowledge of vicriviroc pharmacokinetics in humans, the PK base model was identified by comparing different compartmental models with first-order input and elimination. All inter-individual error terms in the PK parameters were assumed to have a log-normal distribution, i.e.

$$P_j = \theta * \exp(\eta_j)$$

where P is the parameter of interest, j is the jth subject, θ is the estimate of the population mean parameter and ηj is the deviation from the population mean for the jth subject under the assumption that $\eta \sim N(0, \omega_j^2)$.
- Model selection was driven by the data and was based on various goodness of fit indicators, including comparisons based on the Akaike information criteria (AIC), visual inspection of diagnostic scatter plots and evaluation of estimates of population fixed and random effect parameters. Once the random effects covariance matrix was determined, the dataset was examined for outliers by examining weighted residuals.

Covariate Model

- A stepwise forward inclusion procedure (α =0 .01; i.e. the decrease of objective function values larger than 6.63) or using generalized additive modeling (GAM) was performed in PsN or NONMEM to build the full model; and a stepwise backward elimination procedure (α = 0.001 ; i.e. the increase of objective function values larger than 10.83) was applied to determine the final model.

Model Evaluation

- The final model was tested for stability by bootstrap technique. A minimum of 1000 replicates of the data were generated by bootstrap for NONMEM analysis to obtain the mean and standard error of the fixed-effect and random-effect parameters. An internal validation method was also performed by generating a visual predictive check plot of simulated concentrations (mean and 95% prediction interval) overlaid with actual observations to evaluate the predictive performance of the final model. Any problems evident in the simulations was investigated and further model development was conducted as necessary.

Results

Figure 1 Diagnostic Plots

- PK model adequately described the observed data.

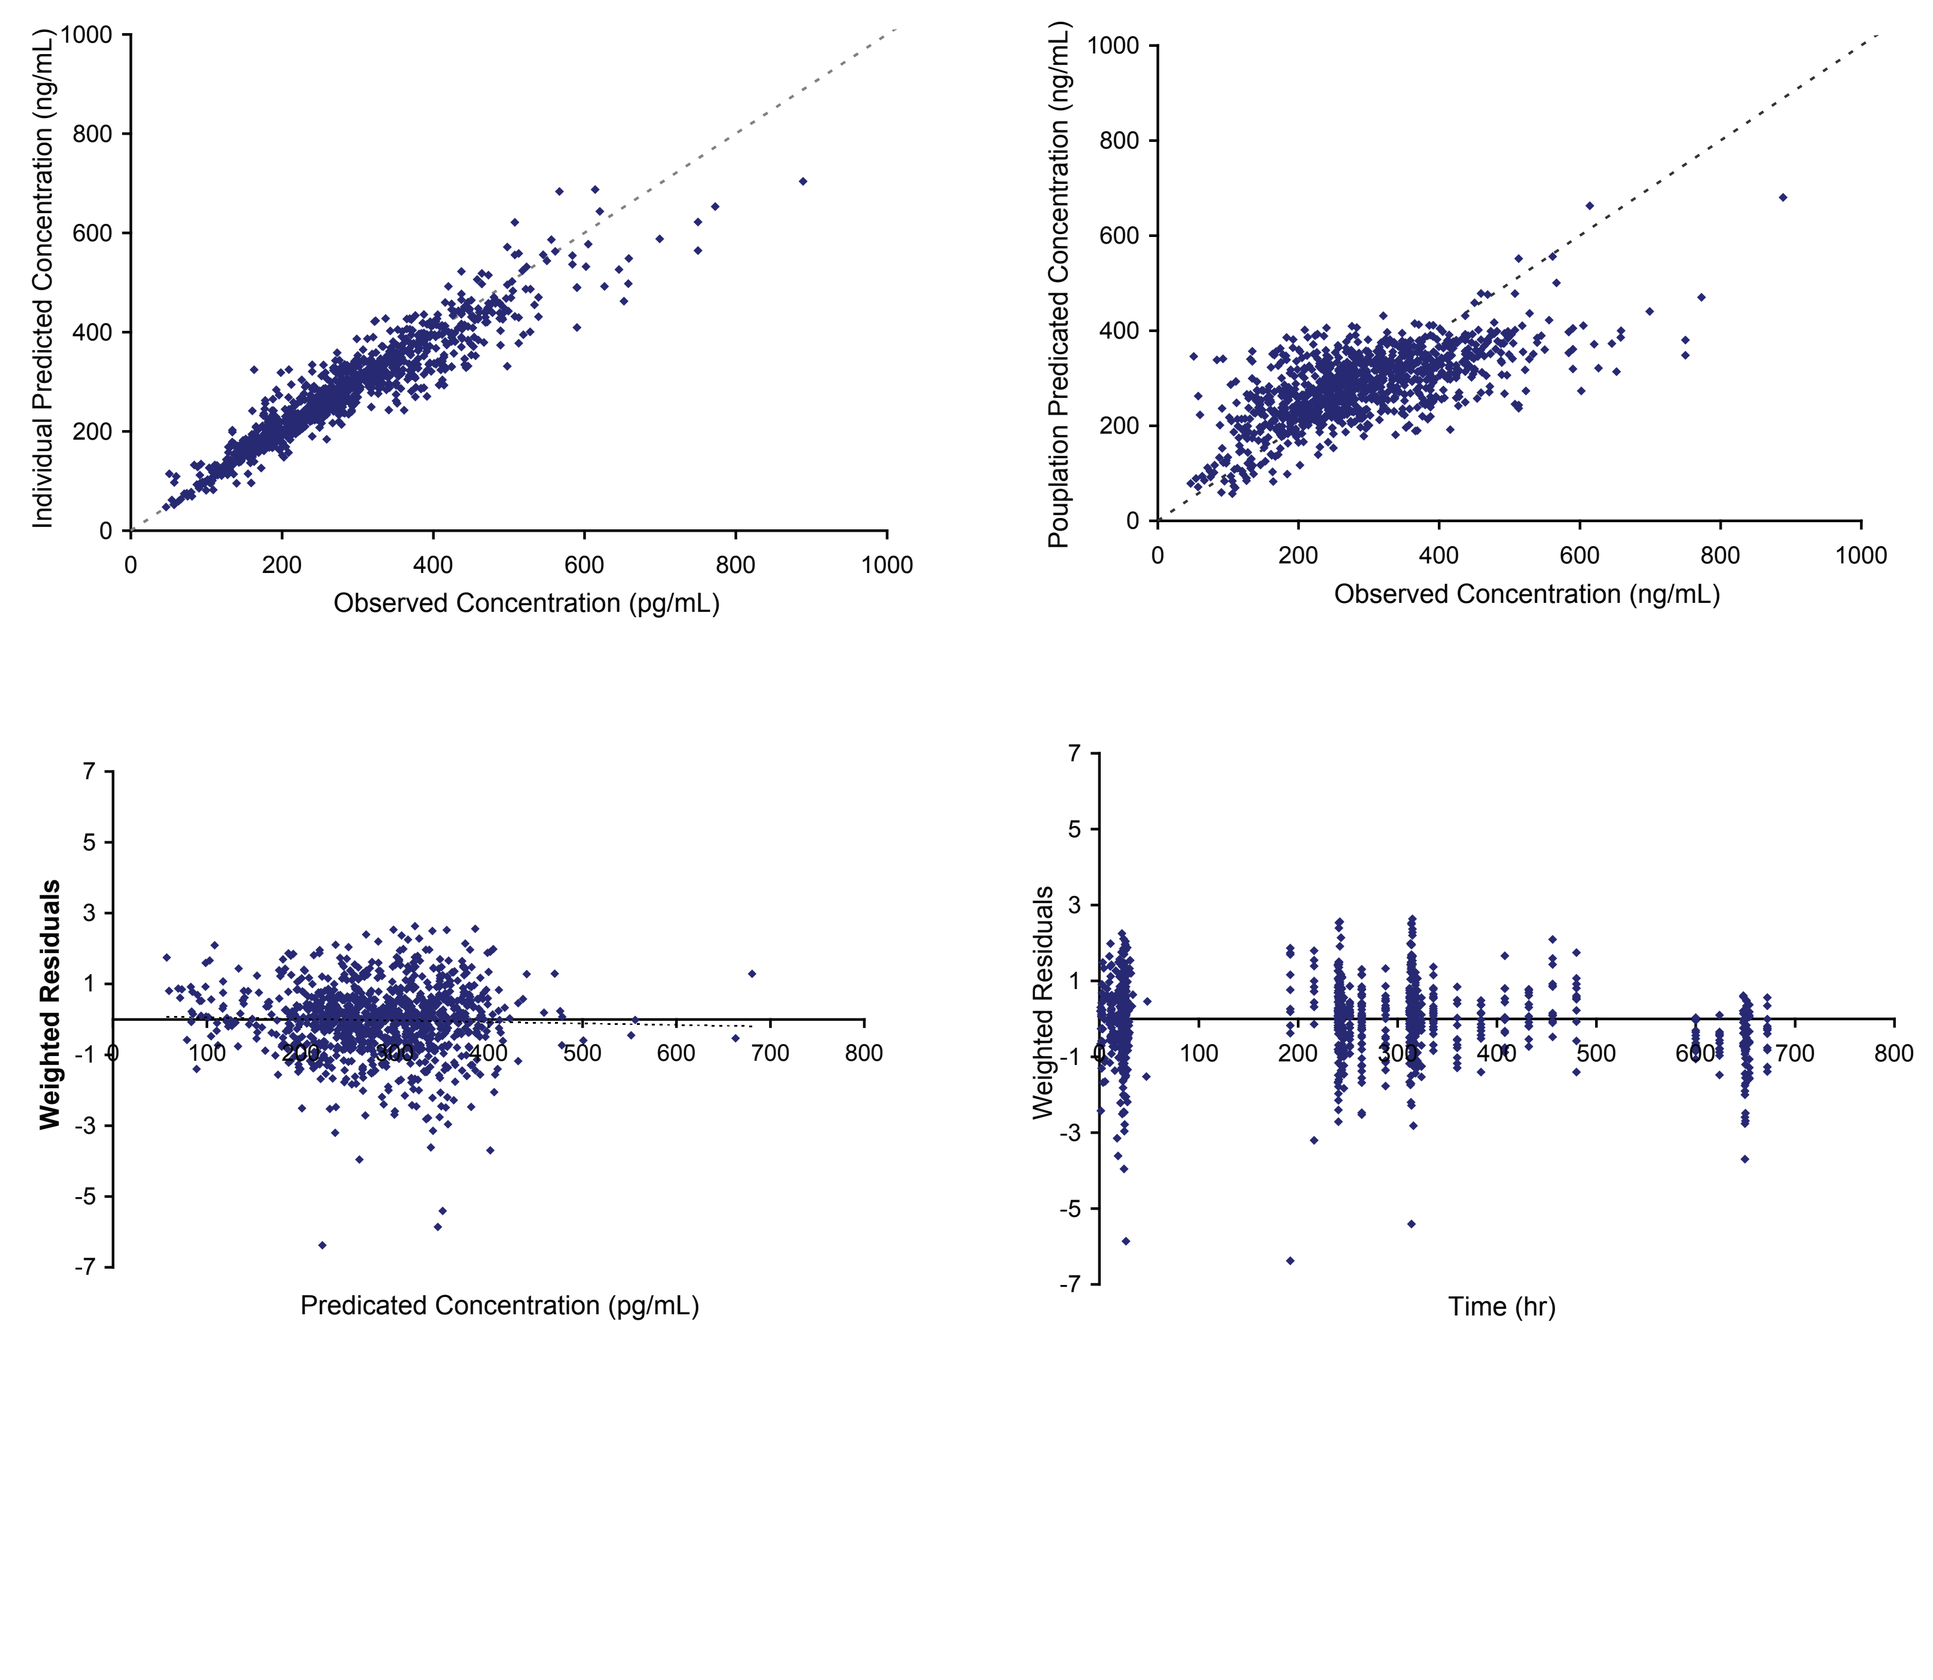

Individual Predicted Concentration (ng/mL)
Observed Concentration (pg/mL)
Pouplation Predicated Concentration (ng/mL)
Observed Concentration (ng/mL)
Weighted Residuals
Predicated Concentration (pg/mL)
Weighted Residuals
Time (hr)

Table 1 Population PK Parameters

- Slow elimination and high volume of distribution
- Significant covariates: Age, Body Weight

Parameters	Estimate (%SE)	Inter-individual variability (%SE)
Ka (1/hr)	0.743 (13)	70.3% (26)
V2/F(L)	140 (9)	40.6% (30)
CL/F (L/hr)	3.39 (2)	29.5% (15)
V3/F(L)	329 (9)	NE
Q/F (L/hr)	26.3 (11)	NE
Covariates	**Estimated Effect (%SE)**	
Body Weight on CL/F	0.787 (23)	
Age on V2/F	0.837 (17)	
Residual variability		
Proportional error1 (HIV)	18.9% (16)	
Proportional error2 (HV)	12.5% (12)	

Ka, absorption rate constant; V_2, volume of distribution at central compartment; CL, total clearance; V_3, volume of distribution at peripheral compartment; Q, intercompartment clearance.

Figure 2 Bootstrap Results

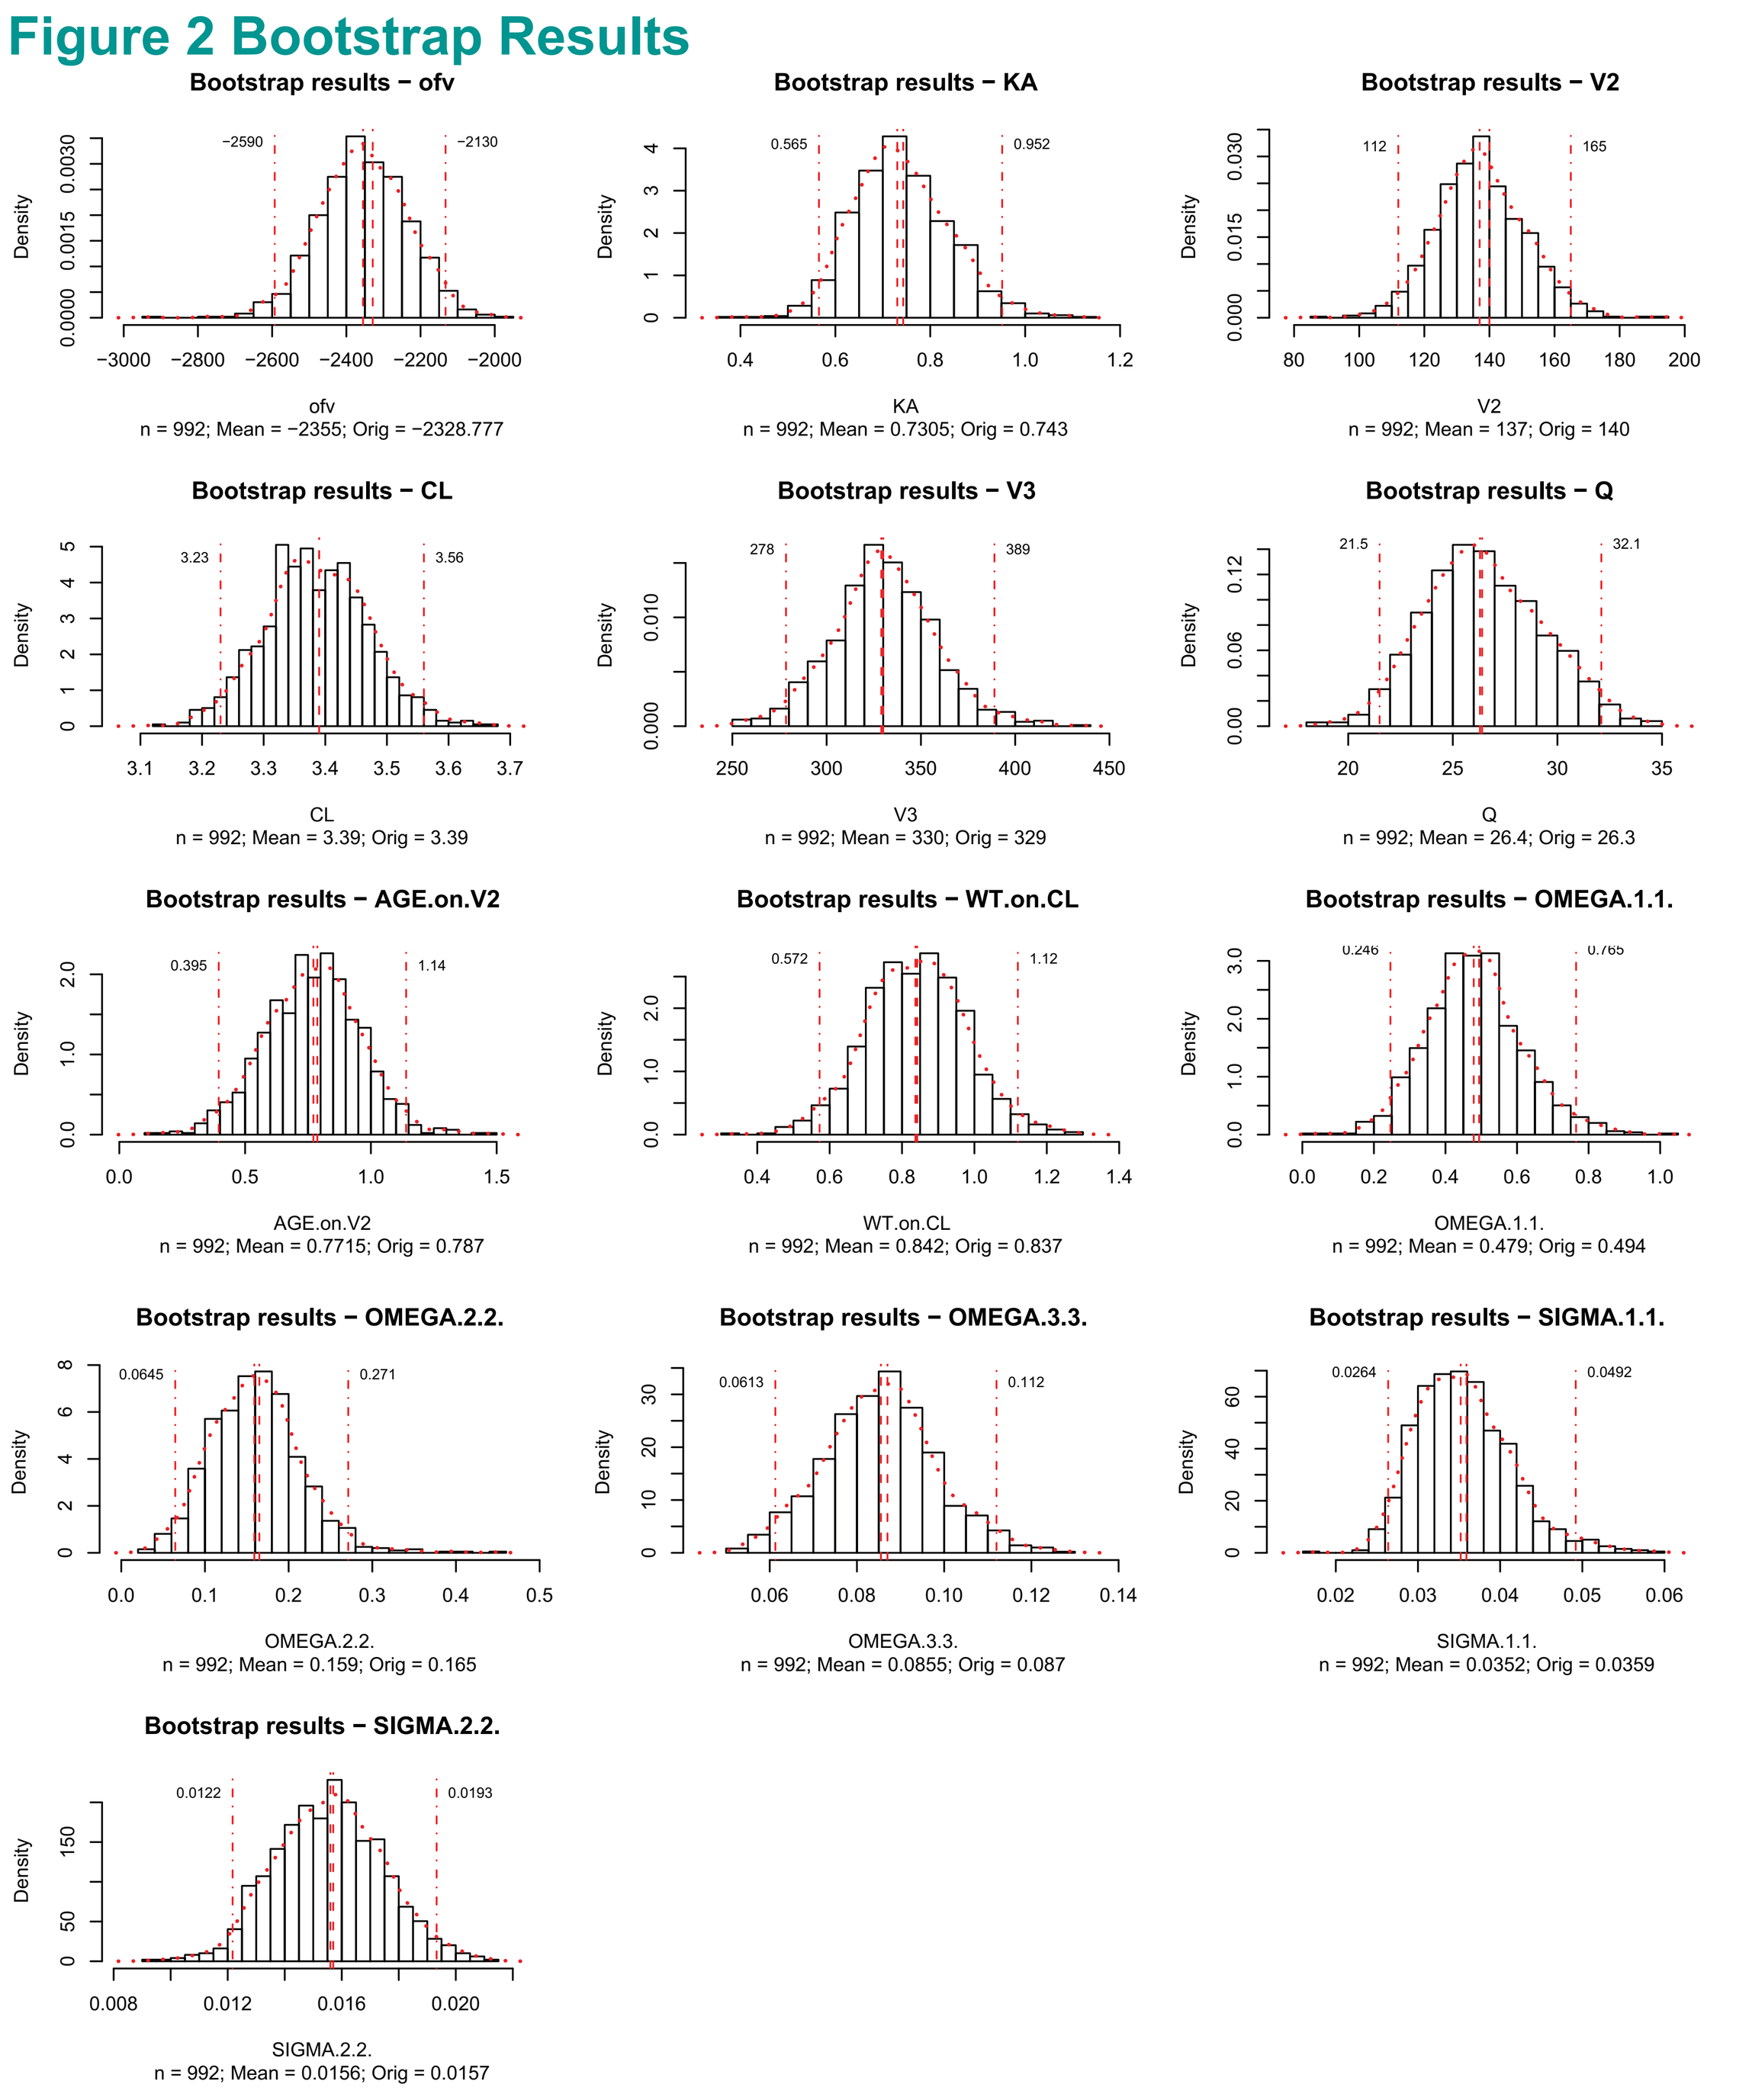

Bootstrap results − ofv
n = 992; Mean = −2355; Orig = −2328.777
Bootstrap results − KA
n = 992; Mean = 0.7305; Orig = 0.743
Bootstrap results − V2
n = 992; Mean = 137; Orig = 140
Bootstrap results − CL
n = 992; Mean = 3.39; Orig = 3.39
Bootstrap results − V3
n = 992; Mean = 330; Orig = 329
Bootstrap results − Q
n = 992; Mean = 26.4; Orig = 26.3
Bootstrap results − AGE.on.V2
n = 992; Mean = 0.7715; Orig = 0.787
Bootstrap results − WT.on.CL
n = 992; Mean = 0.842; Orig = 0.837
Bootstrap results − OMEGA.1.1.
n = 992; Mean = 0.479; Orig = 0.494
Bootstrap results − OMEGA.2.2.
n = 992; Mean = 0.159; Orig = 0.165
Bootstrap results − OMEGA.3.3.
n = 992; Mean = 0.0855; Orig = 0.087
Bootstrap results − SIGMA.1.1.
n = 992; Mean = 0.0352; Orig = 0.0359
Bootstrap results − SIGMA.2.2.
n = 992; Mean = 0.0156; Orig = 0.0157

- The PK parameter estimates from the final model were nearly identical to the respective median values from the bootstrapping runs and the 95% CIs had narrow widths, indicating that the performance and stability of the final population PK model were acceptable.

VPC: Stratify by Disease Status

Figure 3: Visual predictive check with 5th, 50th, and 95th percentiles of simulated and observed vicriviroc concentration (ng/mL) vs time (hr) stratified by disease.

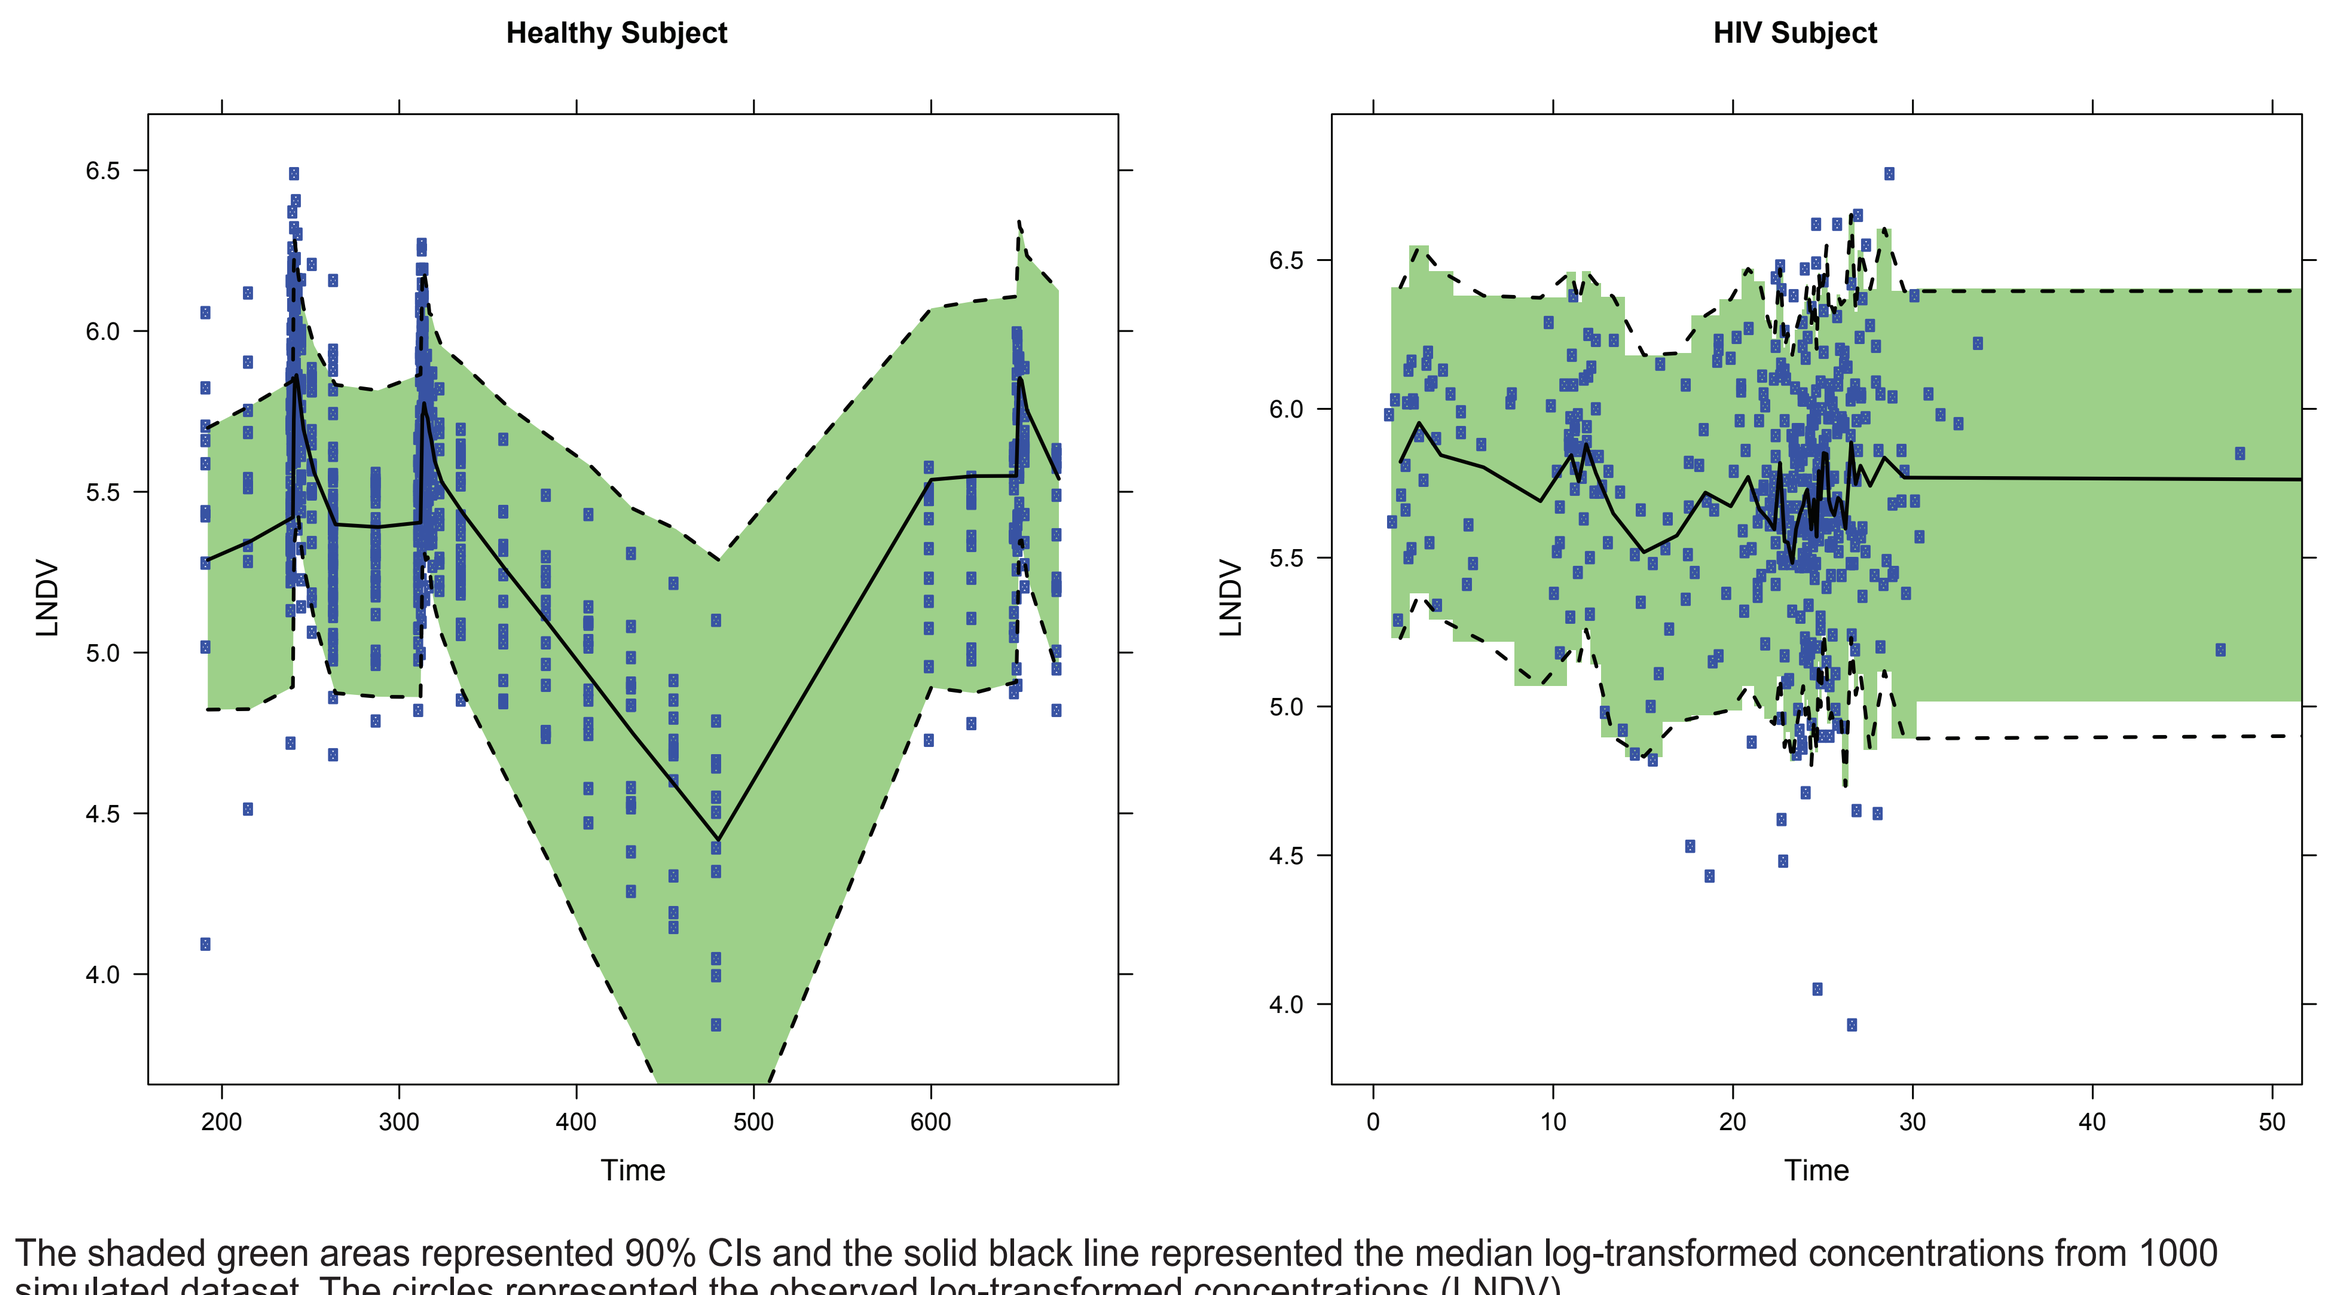

Healthy Subject
HIV Subject
LNDV
Time

The shaded green areas represented 90% CIs and the solid black line represented the median log-transformed concentrations from 1000 simulated dataset. The circles represented the observed log-transformed concentrations (LNDV).

VPC: Stratify by Age

Figure 4: Visual predictive check with 5th, 50th, and 95th percentiles of simulated and observed vicriviroc concentration (ng/mL) vs time (hr) stratified by age

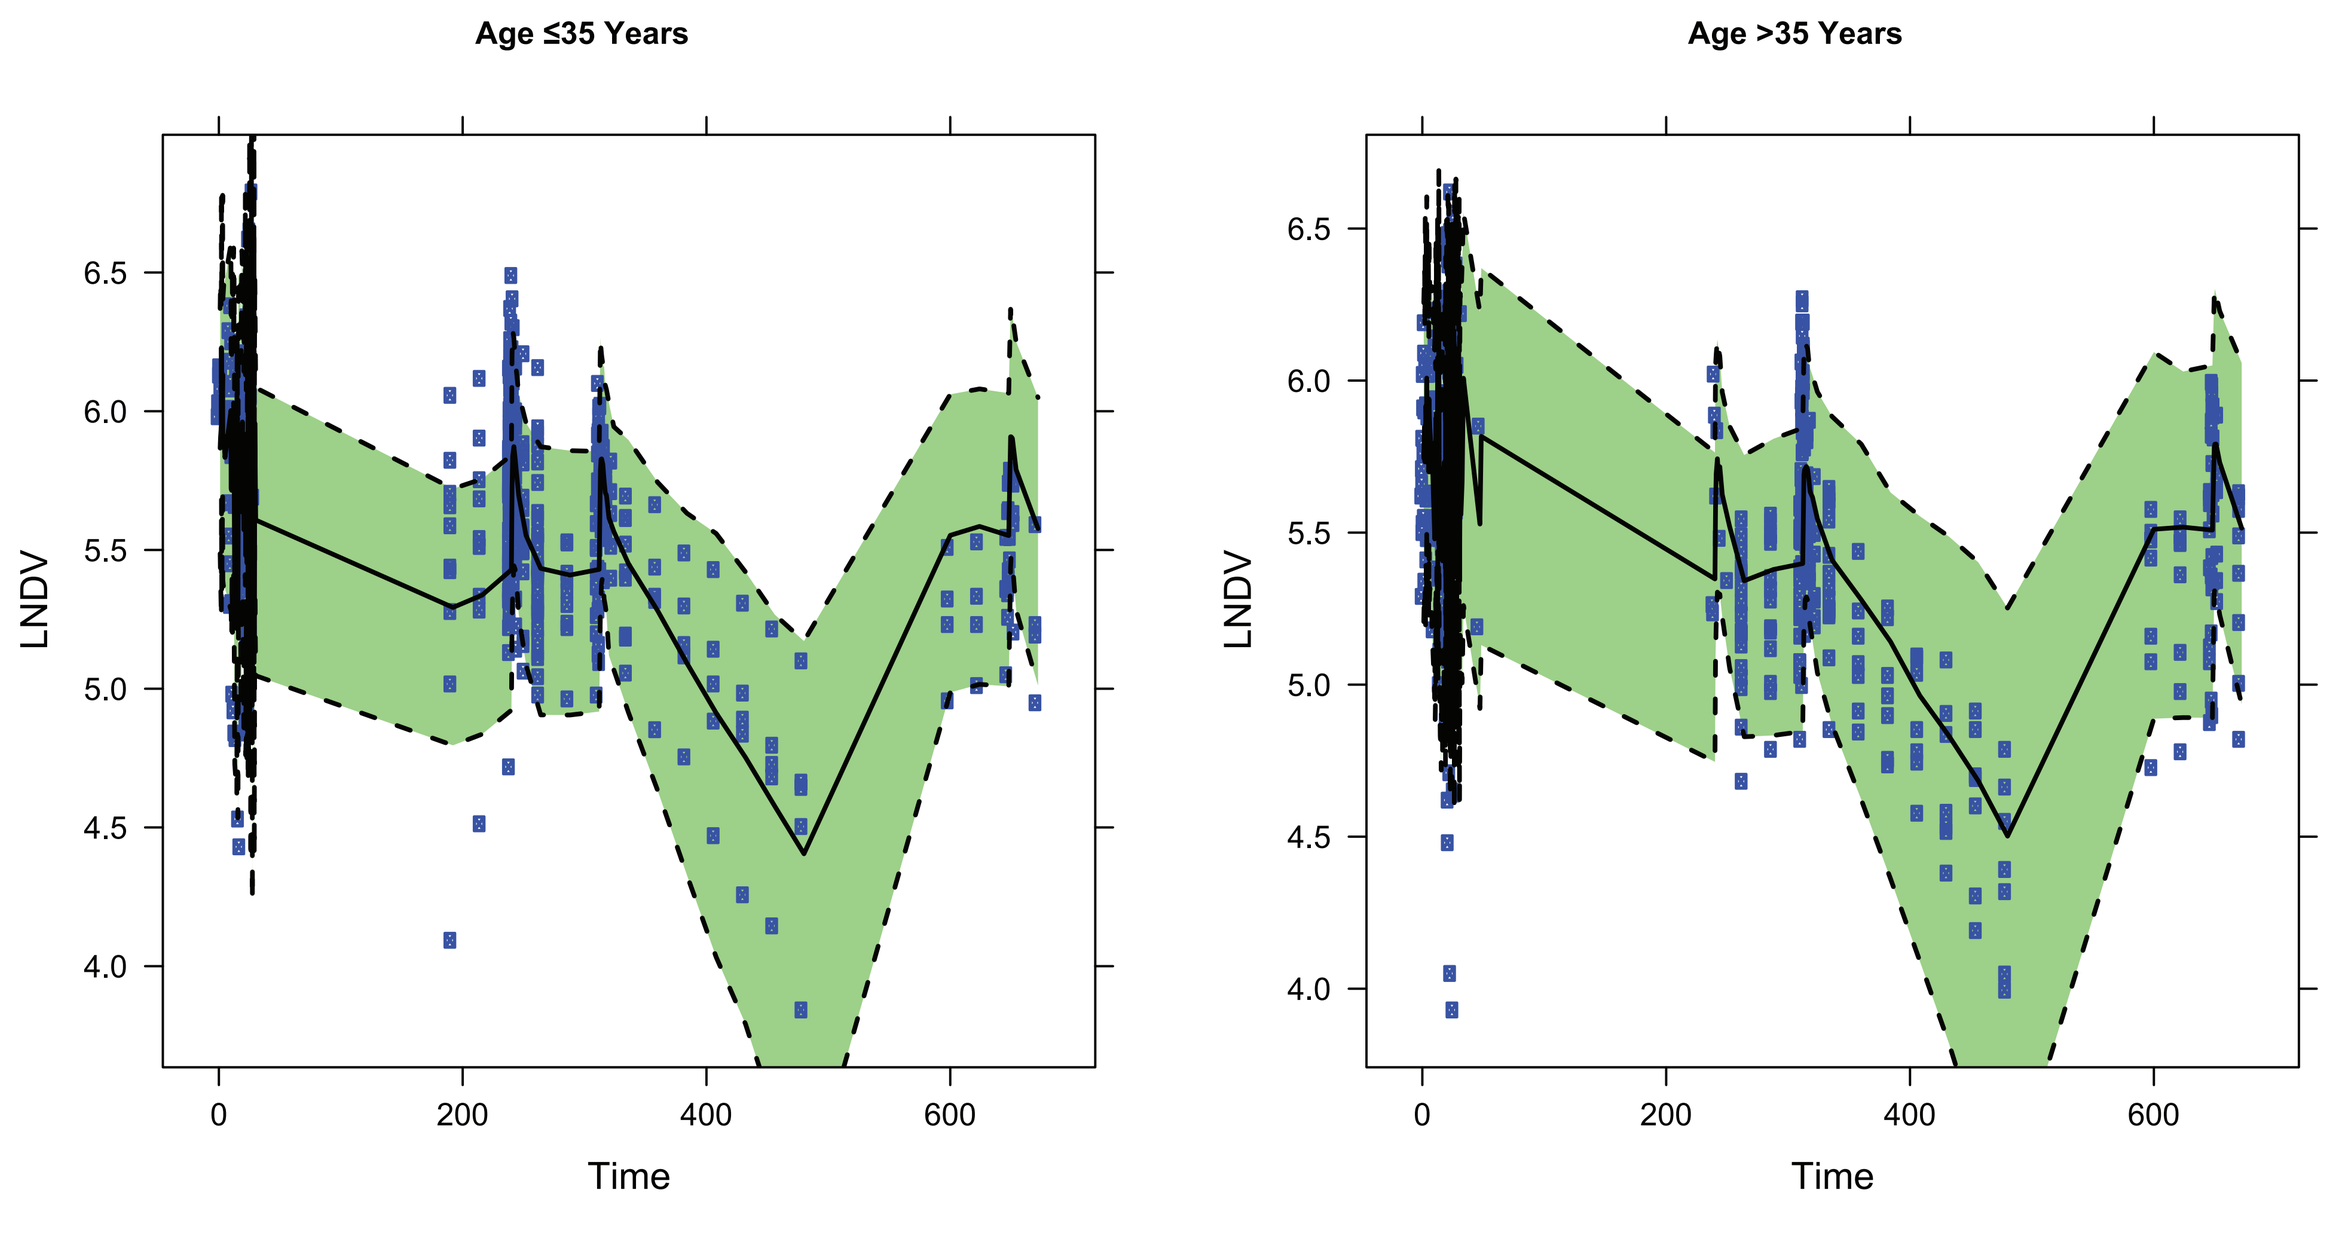

Age ≤35 Years
Age >35 Years
LNDV
Time

The shaded green areas represented 90% CIs and the solid black line represented the median log-transformed concentrations from 1000 simulated dataset. The circles represented the observed log-transformed concentrations (LNDV).

VPC: Stratify by Body Weight

Figure 5: Visual predictive check with 5th, 50th, and 95th percentiles of simulated and observed vicriviroc concentration (ng/mL) vs time (hr) stratified by body weight

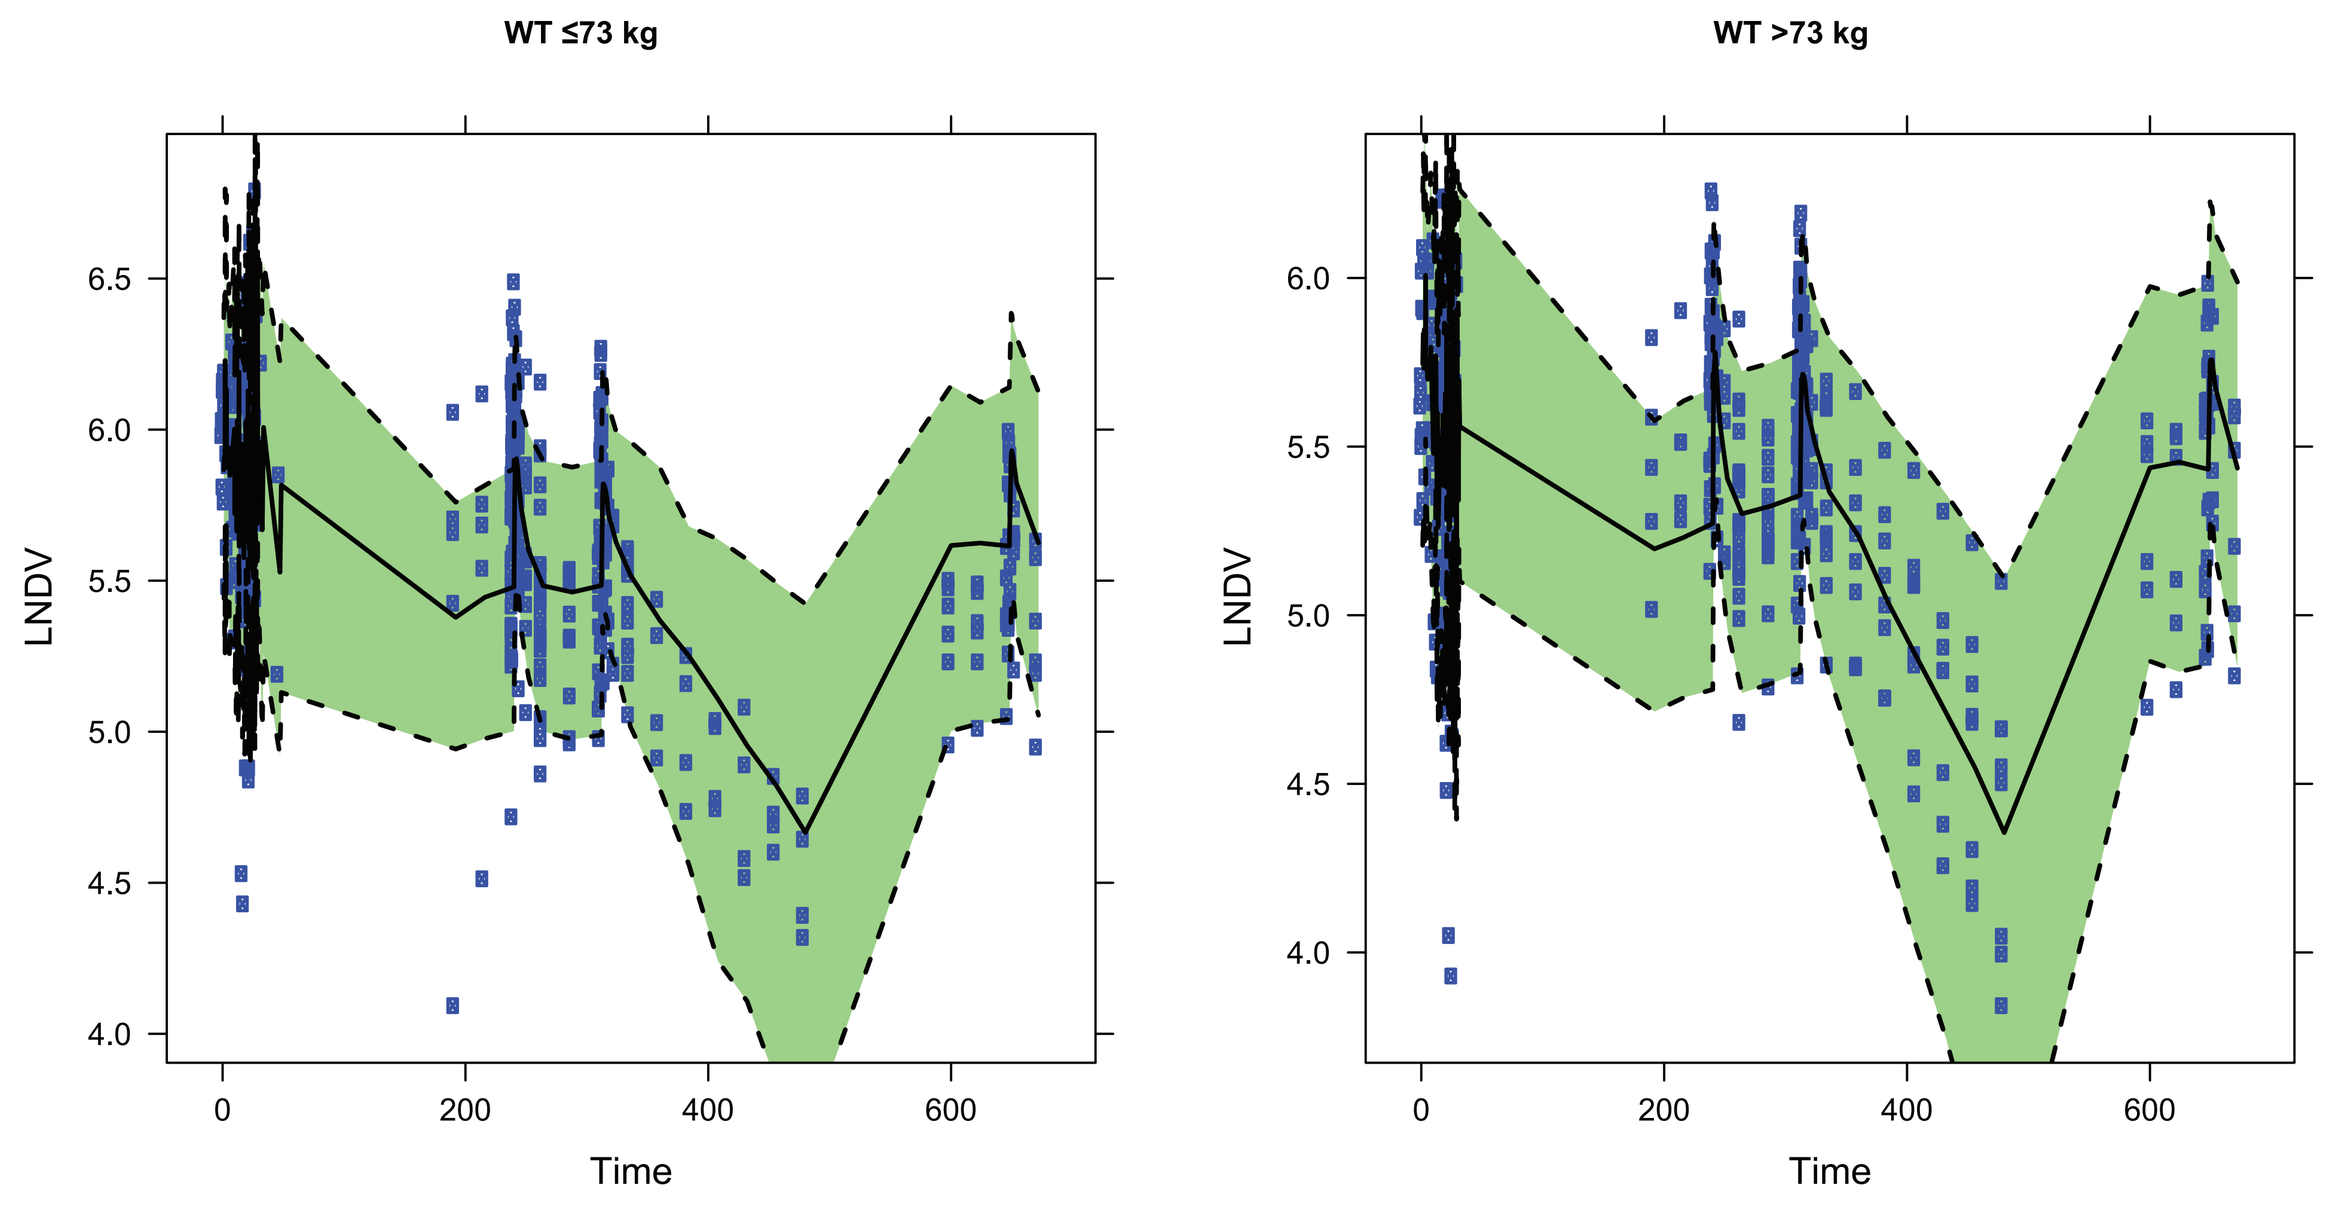

WT ≤73 kg
WT >73 kg
LNDV
Time

The shaded green areas represented 90% CIs and the solid black line represented the median log-transformed concentrations from 1000 simulated dataset. The circles represented the observed log-transformed concentrations (LNDV).

Table 2: Steady state vicriviroc mean pharmacokinetic parameters following 30 mg QD in combination with ritonavir in HIV treatment-naïve subjects.

	Cmin (ng/mL)	Cmax (ng/mL)	AUC (hr*ng/mL)
N		105	
Mean	281	386	7670
SD	98.4	106	2400
CV%	35	27	31
Min	98.7	138	2890
Median	268	373	7360
Max	570	715	14700
Geometric Mean	264	371	7300

Figure 6 PK-PD Relationship

- The correlation between trough drug concentration (Cmin) and viral load change was assessed. Higher response rates were observed in naive subjects with higher vicriviroc Cmin.

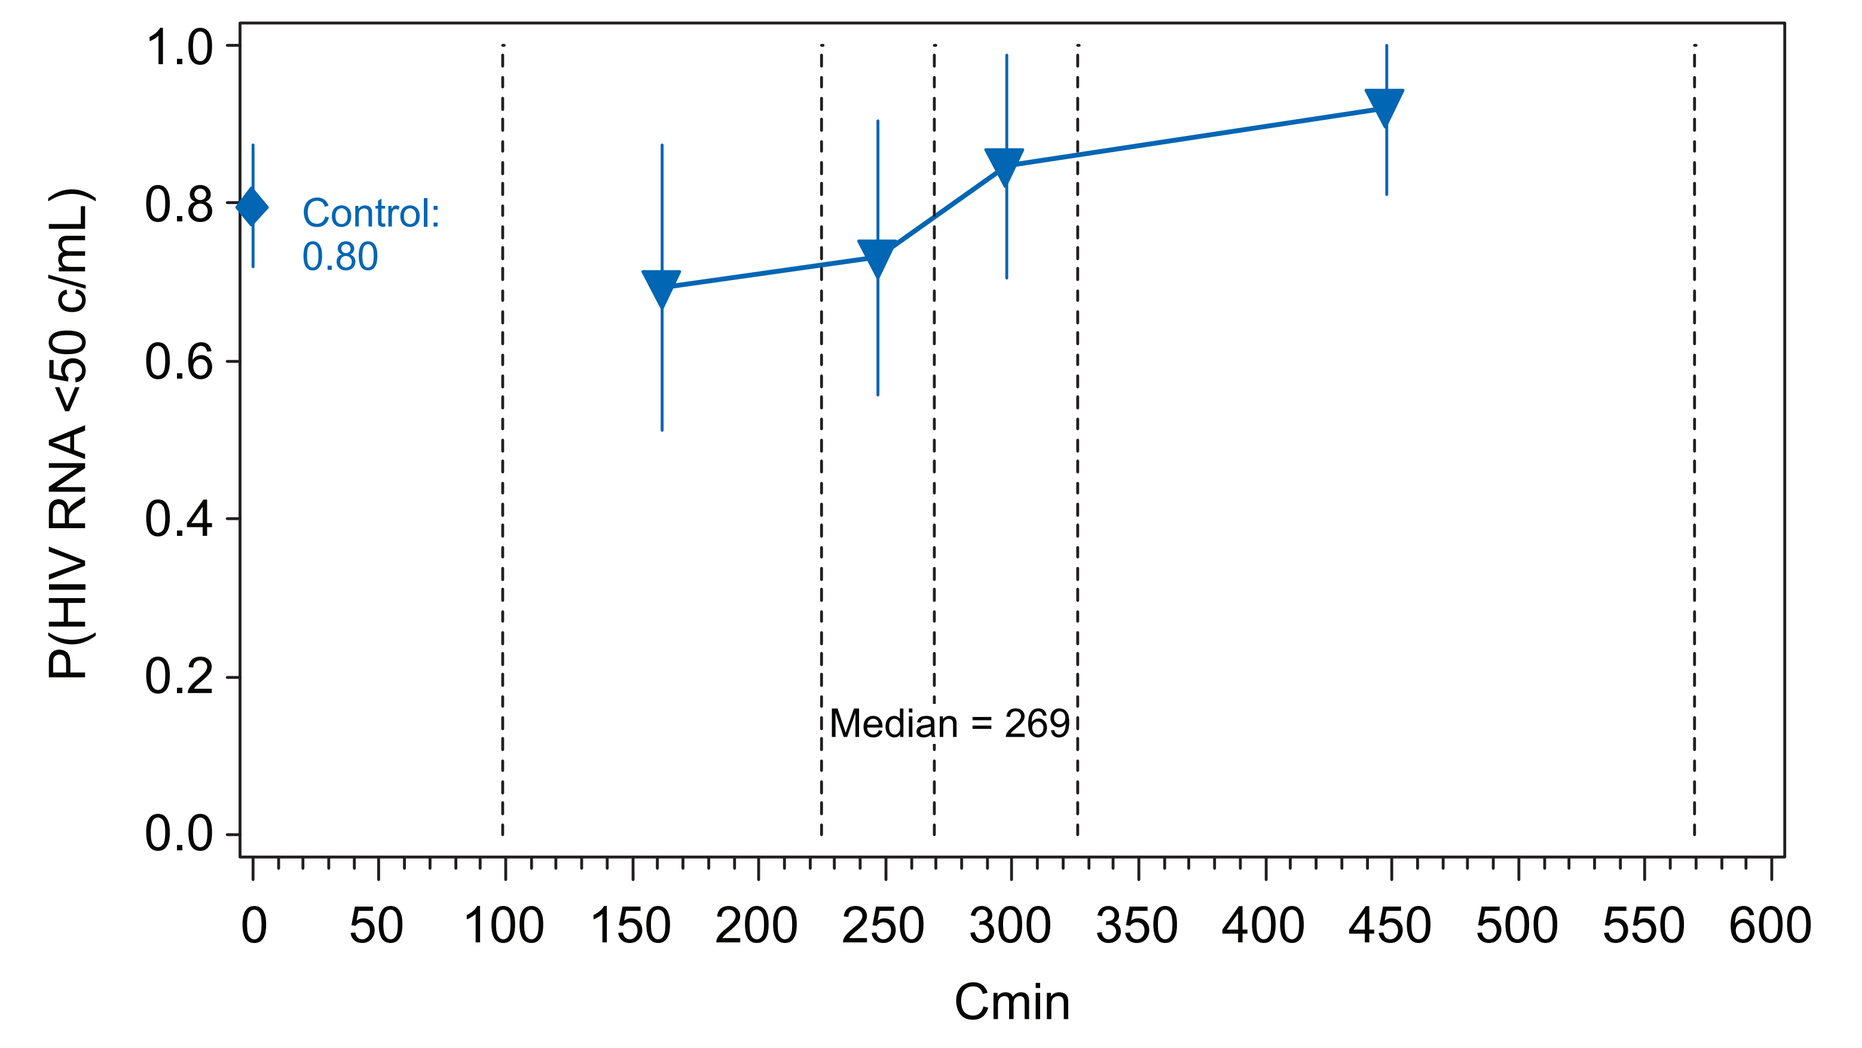

P(HIV RNA <50 c/mL)
Control: 0.80
Median = 269
Cmin

Conclusions

- A two-compartment model adequately described the vicriviroc PK in naïve HIV patients.
- Body weight and age were significant covariates.
- The integrated population PK model and PK-PD association can be used to predict antiviral activity and select the optimal dose regimen in naïve patients.

Copyright © 2011 Merck & Co., Inc., Whitehouse Station, New Jersey, USA, All Rights Reserved

cxu3_201103269_01.indd 5/26/11 Final size: 32"w x 45"h